F-644

LTX-109 - A Rapidly Bactericidal Antimicrobial Drug Highly Effective on Biofilms and CA-MRSA

H. WOLD, B. MORTENSEN, A. FUGELLI, W.M. OLSEN
LYTIX BIOPHARMA, OSLO, NORWAY

Lytix Biopharma

LTX-109

- **A synthetic protein fragment – a peptidomimmetic**
- **High stability against degradation**
- **Produced in large scale**
- **Low cost of synthesis**

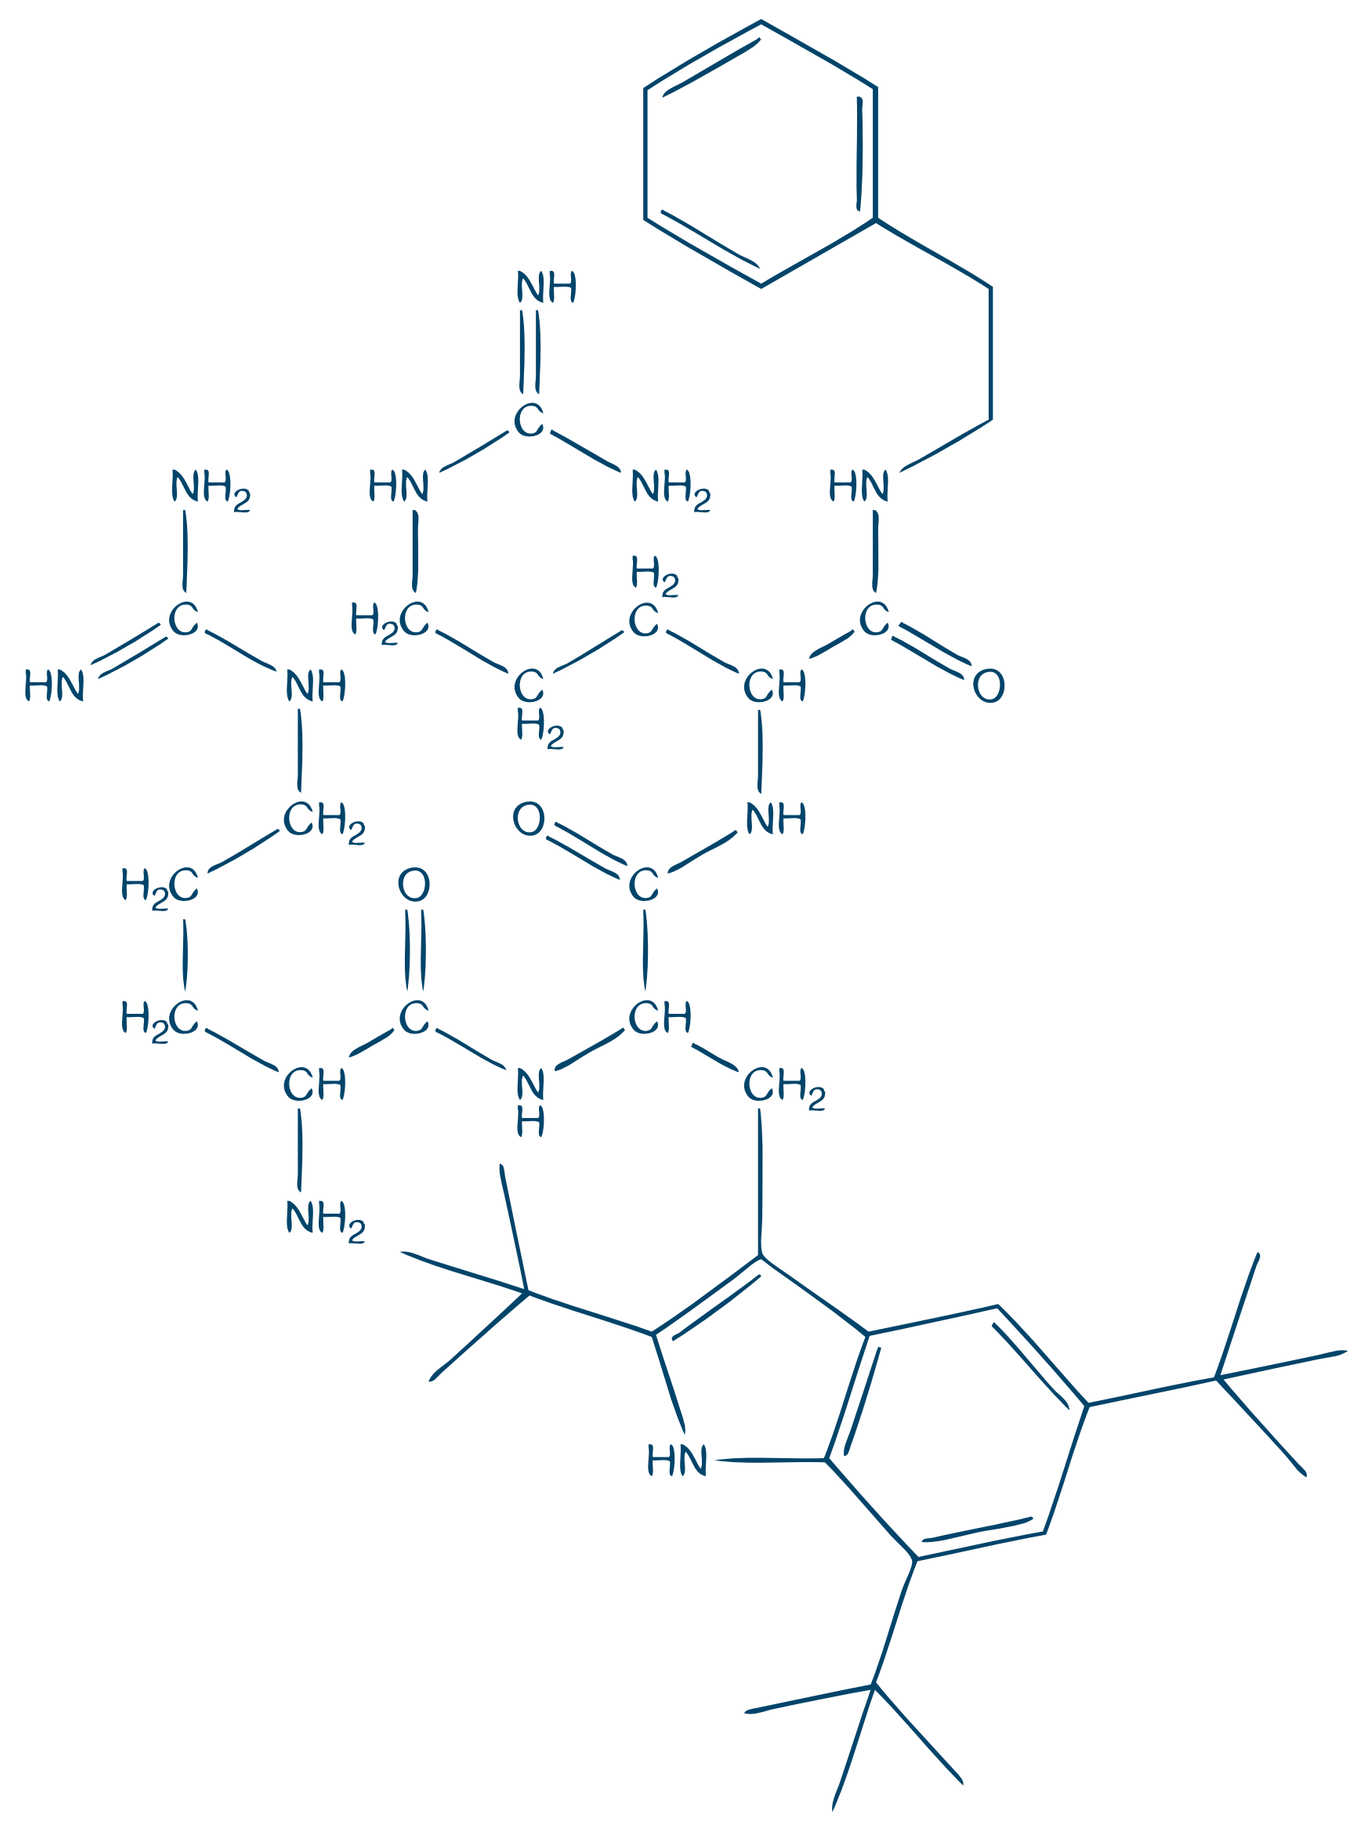

(LTX-109 strukturbilde)

Introduction

Biofilms of staphylococci and other bacterial spices play a role in infections like chronic wounds, chronic otitis media, urinary tract infections, cystic fibrosis, and implant- and catheter associated infections. Such infections affect millions of people each year and many deaths occur as a consequence.

LTX-109 is a novel antimicrobial agent initially being developed as a topical agent for impetigo, nasal decolonisation and biofilm infections. The drug is a mimetic of a membrane-active host defence peptide having a membrane lysing mode of action causing rapid membrane disruption. LTX-109 shows a broad efficacy against a range of pathogens including Gram+ and Gram÷ bacteria, yeasts and fungi.

The compound is equally effective against antibiotic-resistant species such as methicillin resistant *Staphylococcus aureus* (MRSA), vancomycin resistant *Enterococci* (VRE) and multi-resistant *Pseudomonas* isolates. LTX-109 shows a low propensity for resistant development and no in vitro cross-resistance with other classes of antibiotics.

Objective

There is a medical need for new and fast-acting antimicrobials with activity against bacterial biofilms. The present studies were designed to investigate the potential effects of LTX-109 in two MRSA skin biofilm models in mice. in both models the effect of LTX-109 was compare with mupirocin, the “gold standard” for skin infections.

Methods

The two different biofilm models are as described under «Results».

Results

LTX-109 demonstrates a significantly faster reduction of CA-MRSA (USA300) tissue load compared to mupirocin

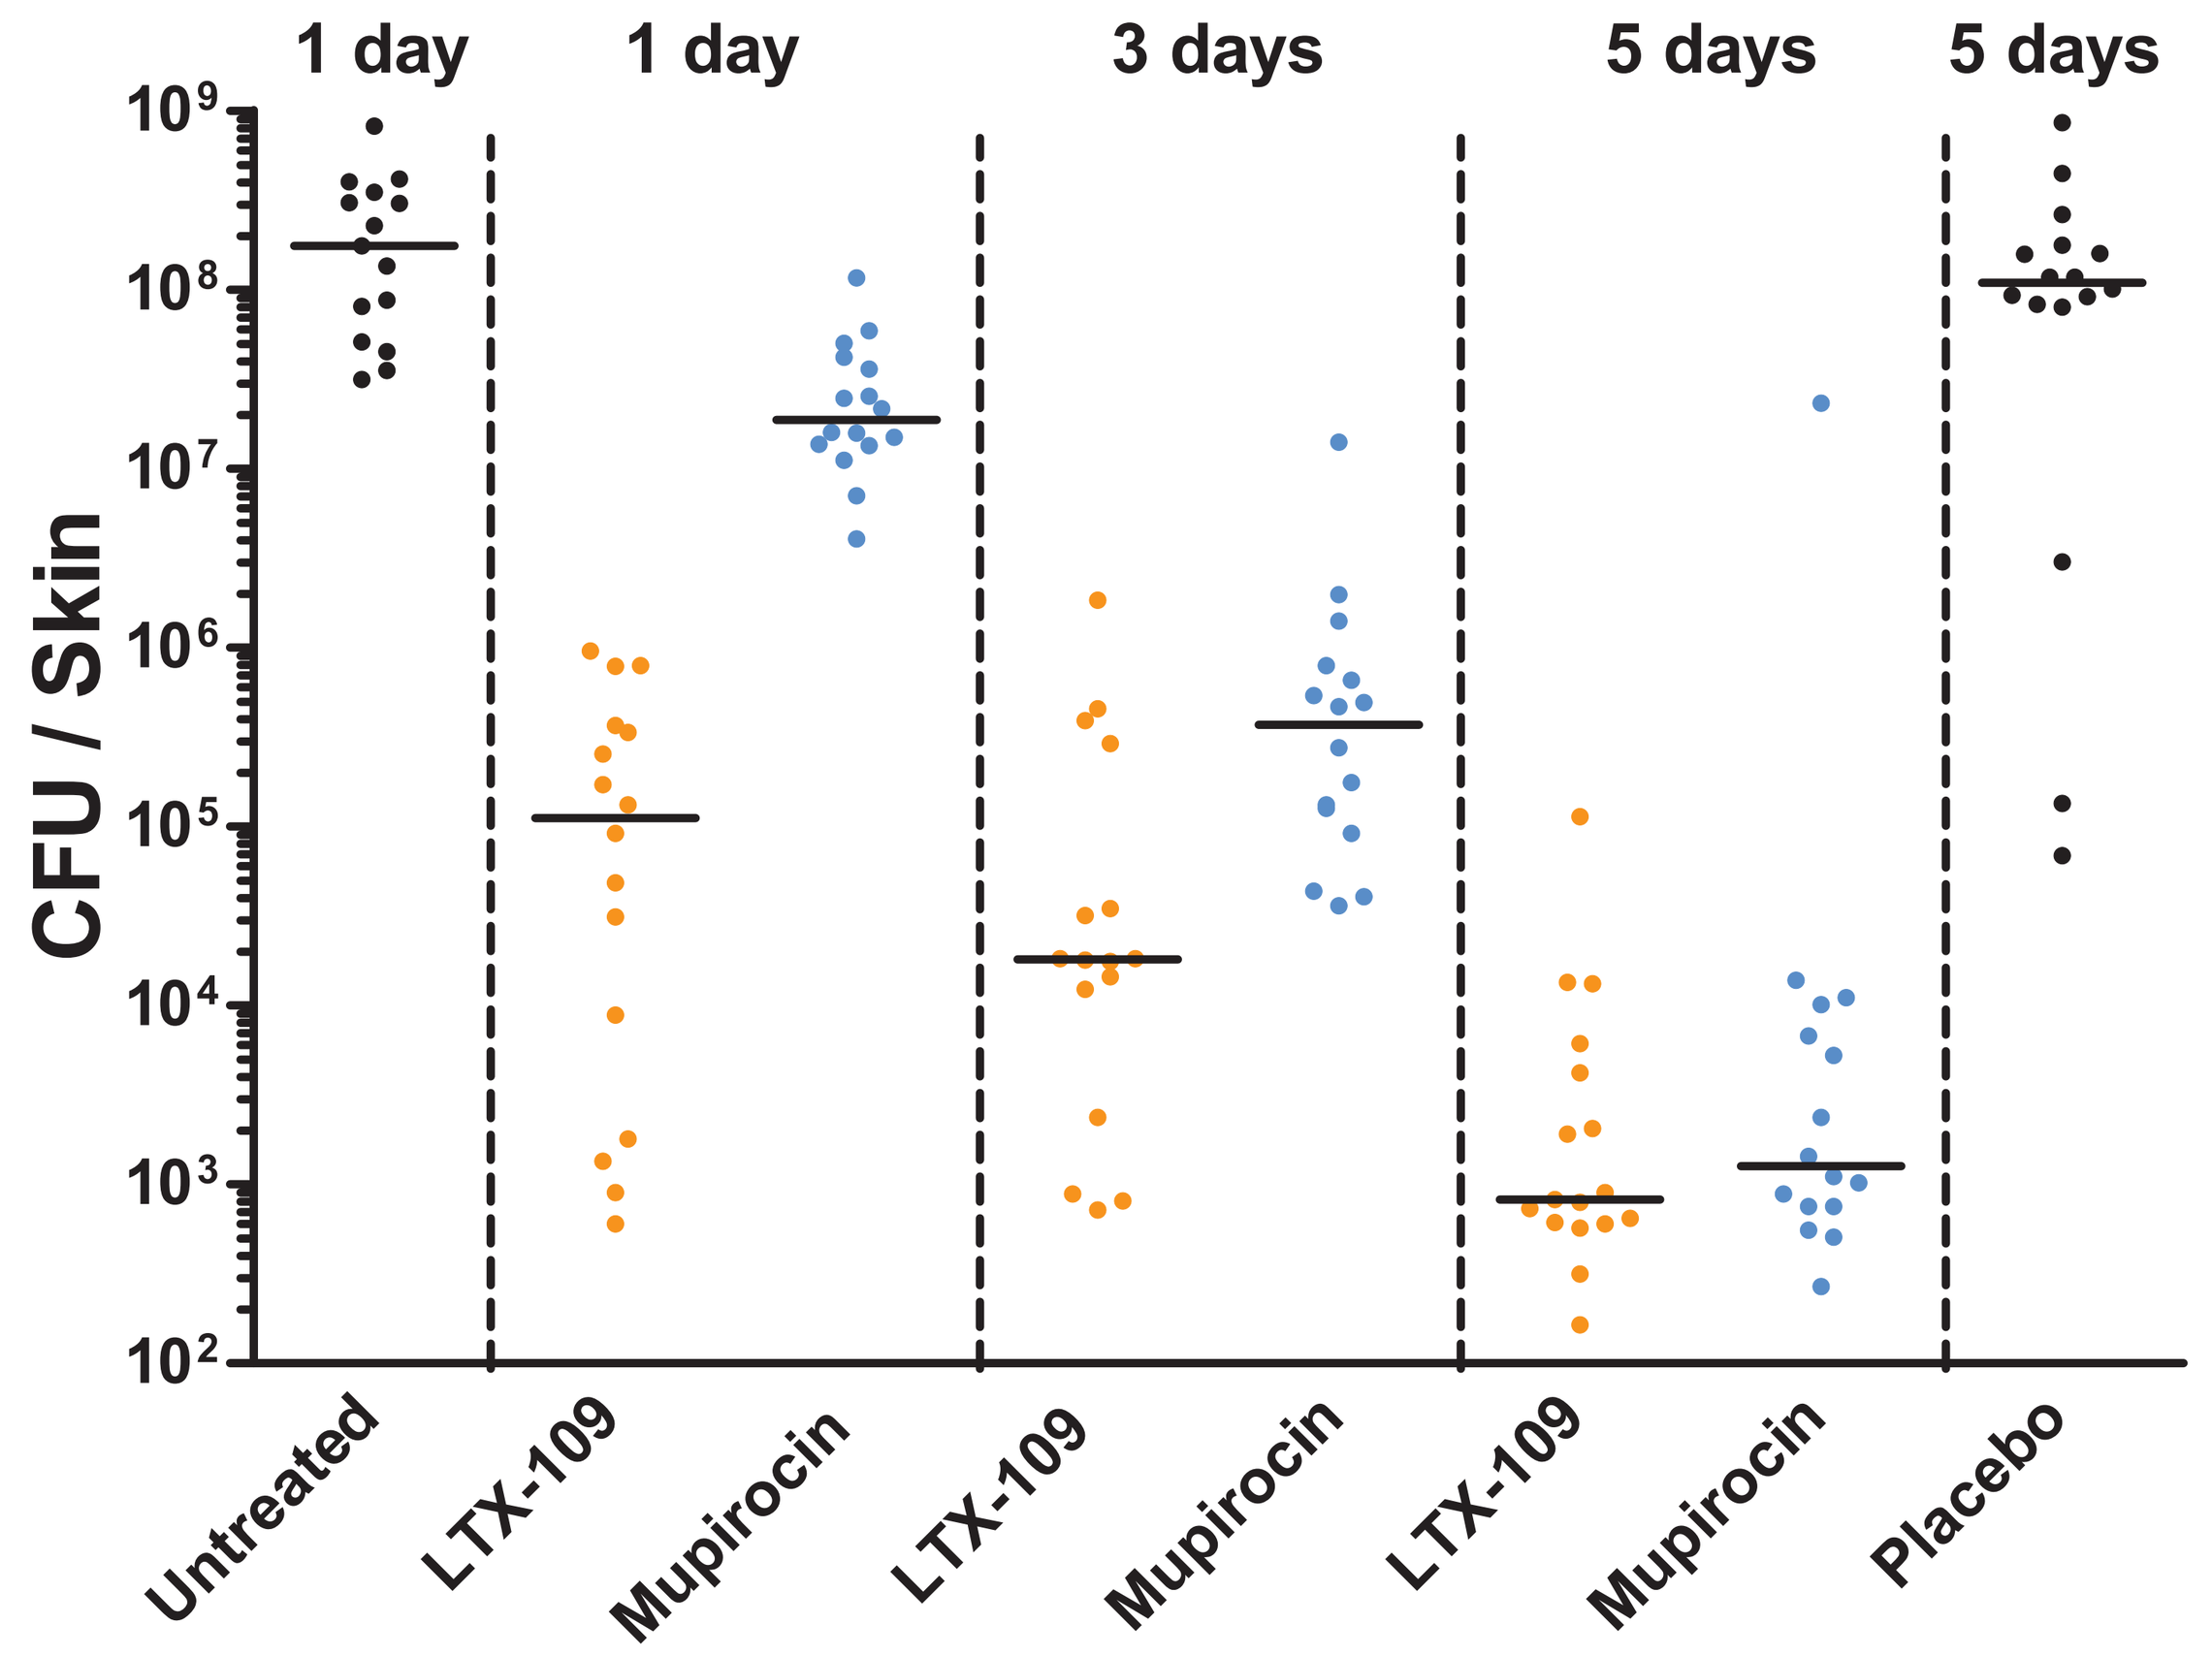
1 day
1 day
3 days
5 days
5 days
CFU / Skin
10^9
10^8
10^7
10^6
10^5
10^4
10^3
10^2
Untreated
LTX-109
Mupirocin
LTX-109
Mupirocin
LTX-109
Mupirocin
Placebo

Infection was established after tape-stripping and scalpel blade-cut injury of mouse skin followed by inoculation with suspensions of community acquired MRSA (CA-MRSA) (USA 300). The next day mice were treated TID (every 3 hours) with 2 % LTX-109, 2 % mupirocin or placebo for either 1, 3 or 5 days. Bacterial tissue load was determined by colony forming units (CFU) from skin biopsy samples.

- LTX-109 treatment for 1 and 3 days was highly effective and significantly better in reducing CA-MRSA tissue load compared to mupirocin
- 5 days treatment with mupirocin was needed to achieve as good effect as with LTX-109

LTX-109 demonstrates a significant effect on MRSA wound biofilm

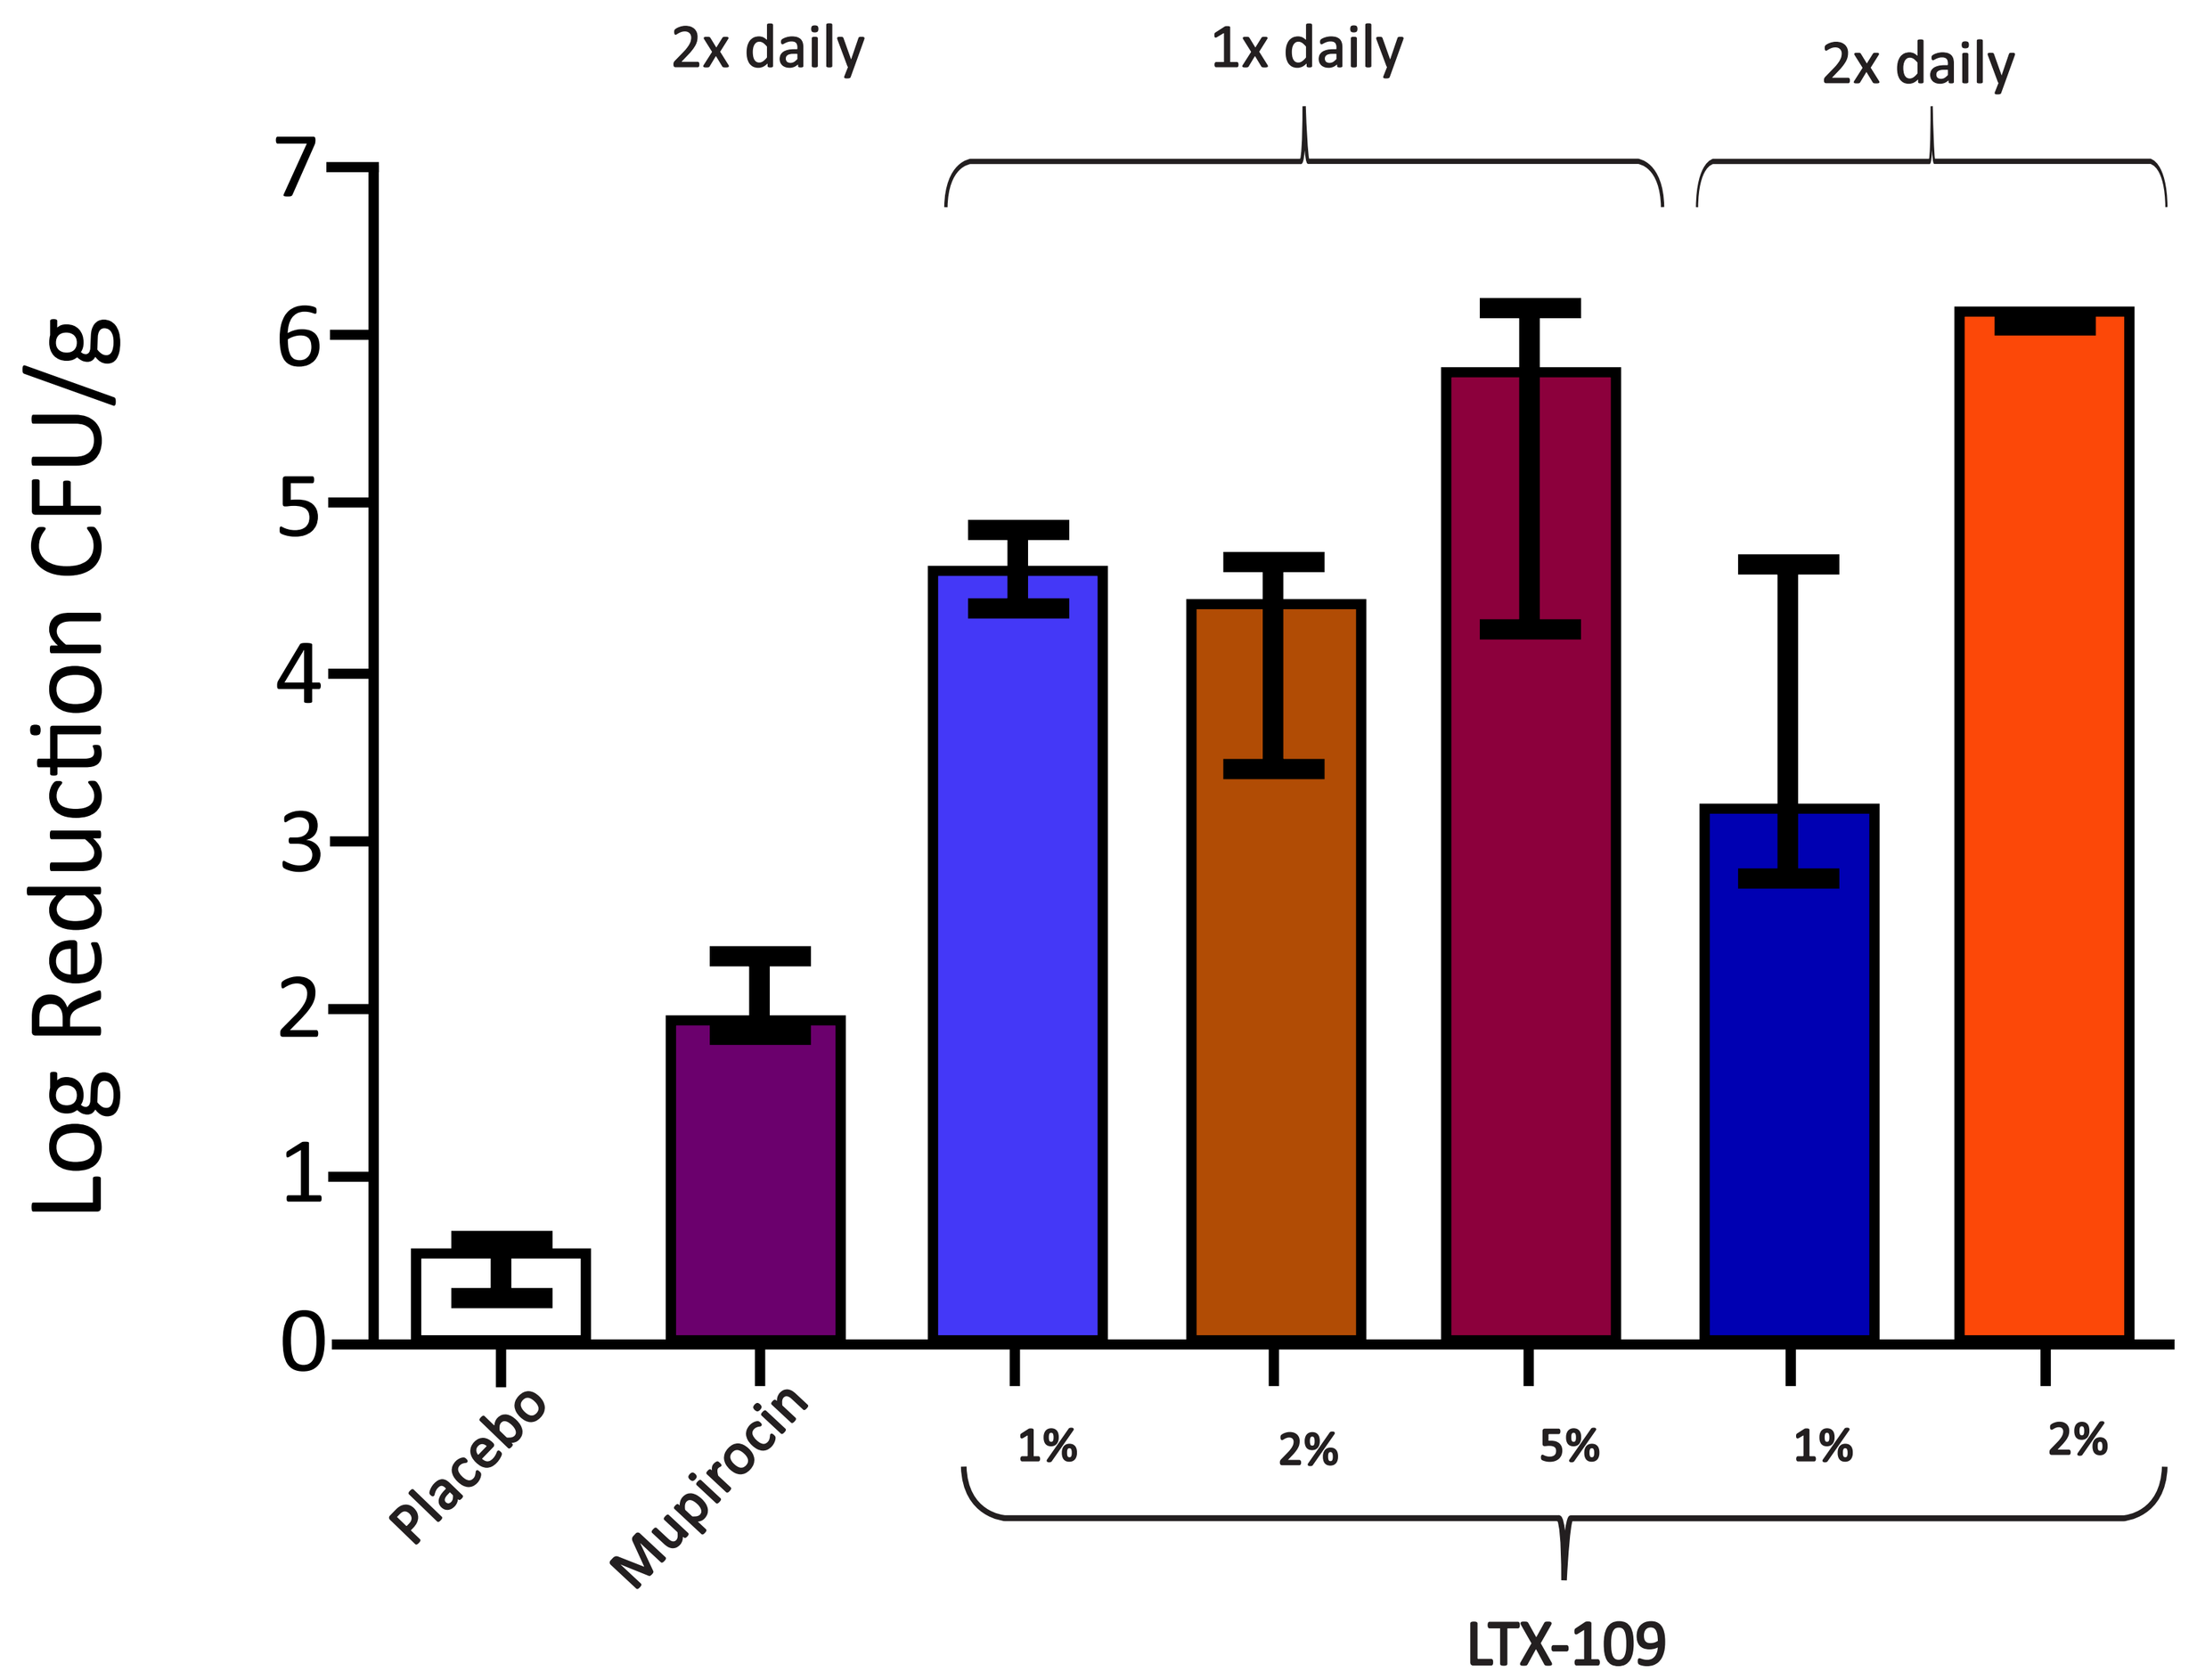
2x daily
1x daily
2x daily
Log Reduction CFU/g
7
6
5
4
3
2
1
0
Placebo
Mupirocin
1%
2%
5%
1%
2%
LTX-109

Partial thickness wounds were established at the dorsal skin of mice using a sanding tool. MRSA biofilm were allowed to establish over 24 hours. Wounds were treated 1 or 2 times daily (every 8 hours) with 1 %, 2 % or 5 % LTX-109 or with mupirocin 2 times daily for 2 days. Moist occlusion was used to cover.

- Once and twice daily treatment of MRSA wound biofilm with 1, 2 or 5 % LTX-109 were better than 2 times treatment with mupirocin
- LTX-109 treatment once daily showed median reductions in bioburden of > 4 log CFU/g relative to control, whereas mupirocin achieved a 1.9 log CFU/g reduction relative to control

Conclusions

- Today there is an unmet medical need for a novel and effective drug with low propensity for resistance development for treatment of biofilm infections, including those caused by nosocomial or community acquired MRSA
- The effect of LTX-109 on reducing bacterial tissue load was significantly better after 1 and 3 days treatment compared to mupirocin, indicating a faster resolution of the infection with LTX-109
- The wound model also demonstrated that both once and twice daily treatment with LTX-109 were better than mupirocin. These data indicates that even lower doses of LTX-109 could be effective for treating biofilm infections.
- The present models demonstrate the potential utility of LTX-109 as fast acting bactericidal drug in treatment of biofilms and skin infections, even those caused by MRSA

LTX-109

- Novel mechanism of action and broad spectrum of activity
- Low propensity for resistance development and active against drug-resistant strains
- Effective against fungal and bacterial biofilms
- Superior efficacy compared to market leaders (Bactroban®, Fucidin®, Altabax®/ Altargo®)
- In Phase I and two Phase I/IIa trials LTX-109 has demonstrated good tolerance and minimal systemic bioavailability
- LTX-109 has demonstrated Proof-of-Concept in decolonisation of nasal MRSA/ MSSA.
- Currently a Phase II Proof-of-Concept study in a large population with impetigo is ongoing, where results are expected in first half of 2014

© WWW.CHILIGROUP.NO|2013

Lytix Biopharma AS | P.O. Box 6447 | NO-9294 Tromsø, Norway | E-mail: post@lytixbiopharma.com | Phone: +47 77 67 55 00 | Fax: +47 77 67 55 01