A phase I, dose escalation study of LTX-315 as monotherapy or in combination with either ipilimumab or pembrolizumab, in patients with transdermally accessible tumors (NCT01986426)

CT108

JEAN-FRANCOIS BAURAIN[1], AHMAD AWADA[2], PAAL F. BRUNSVIG[3], REBECCA KRISTELEIT[4], DAG ERIK JØSSANG[5], NINA LOUISE JEBSEN[5], AURÉLIEN MARABELLE[6], DELPHINE LOIRAT[7], LAURENCE ZITVOGEL[6], GUIODO KROEMER[6], BALDUR SVEINBJØRNSSON[8], ØYSTEIN REKDAL[8], ANDREW SAUNDERS[8], BERIT NICOLAISEN[8], HEDDA WOLD[8] AND JAMES SPICER[9]

[1]Saint-Luc University Hospital Brussels, Université catholique de Louvain, Belgium, [2]Institut Jules Bordet, Université Libre de Bruxelles, Belgium, [3]Oslo University Hospital, Norway, [4]University College London Hospital, UK, [5]Haukeland University Hospital, Bergen, Norway, [6]Institut Gustave Roussy, Paris, France, [7]Institut Curie, Paris, France [8]Lytix Biopharma AS [9]King's College London, Guy's Hospital, UK.

Lytix Biopharma

Background

LTX-315, a first in class oncolytic peptide is developed from host defense peptides that have an important function in innate immune responses to microbial pathogens (1).

Pre-clinical studies of LTX-315 demonstrates:

- Unique immunogenic cell death
 - Targets and disintegrates intracellular organelles
 - Release of potent immune stimulating molecules and tumor antigens (2)
- Increase infiltration of CD8+ T cells in both injected and non-injected tumors (3)
- Reduce number of Tregs and MDSC in injected tumors
- Systemic tumor specific immune responses

Unique mode of action "Release and Reshape"

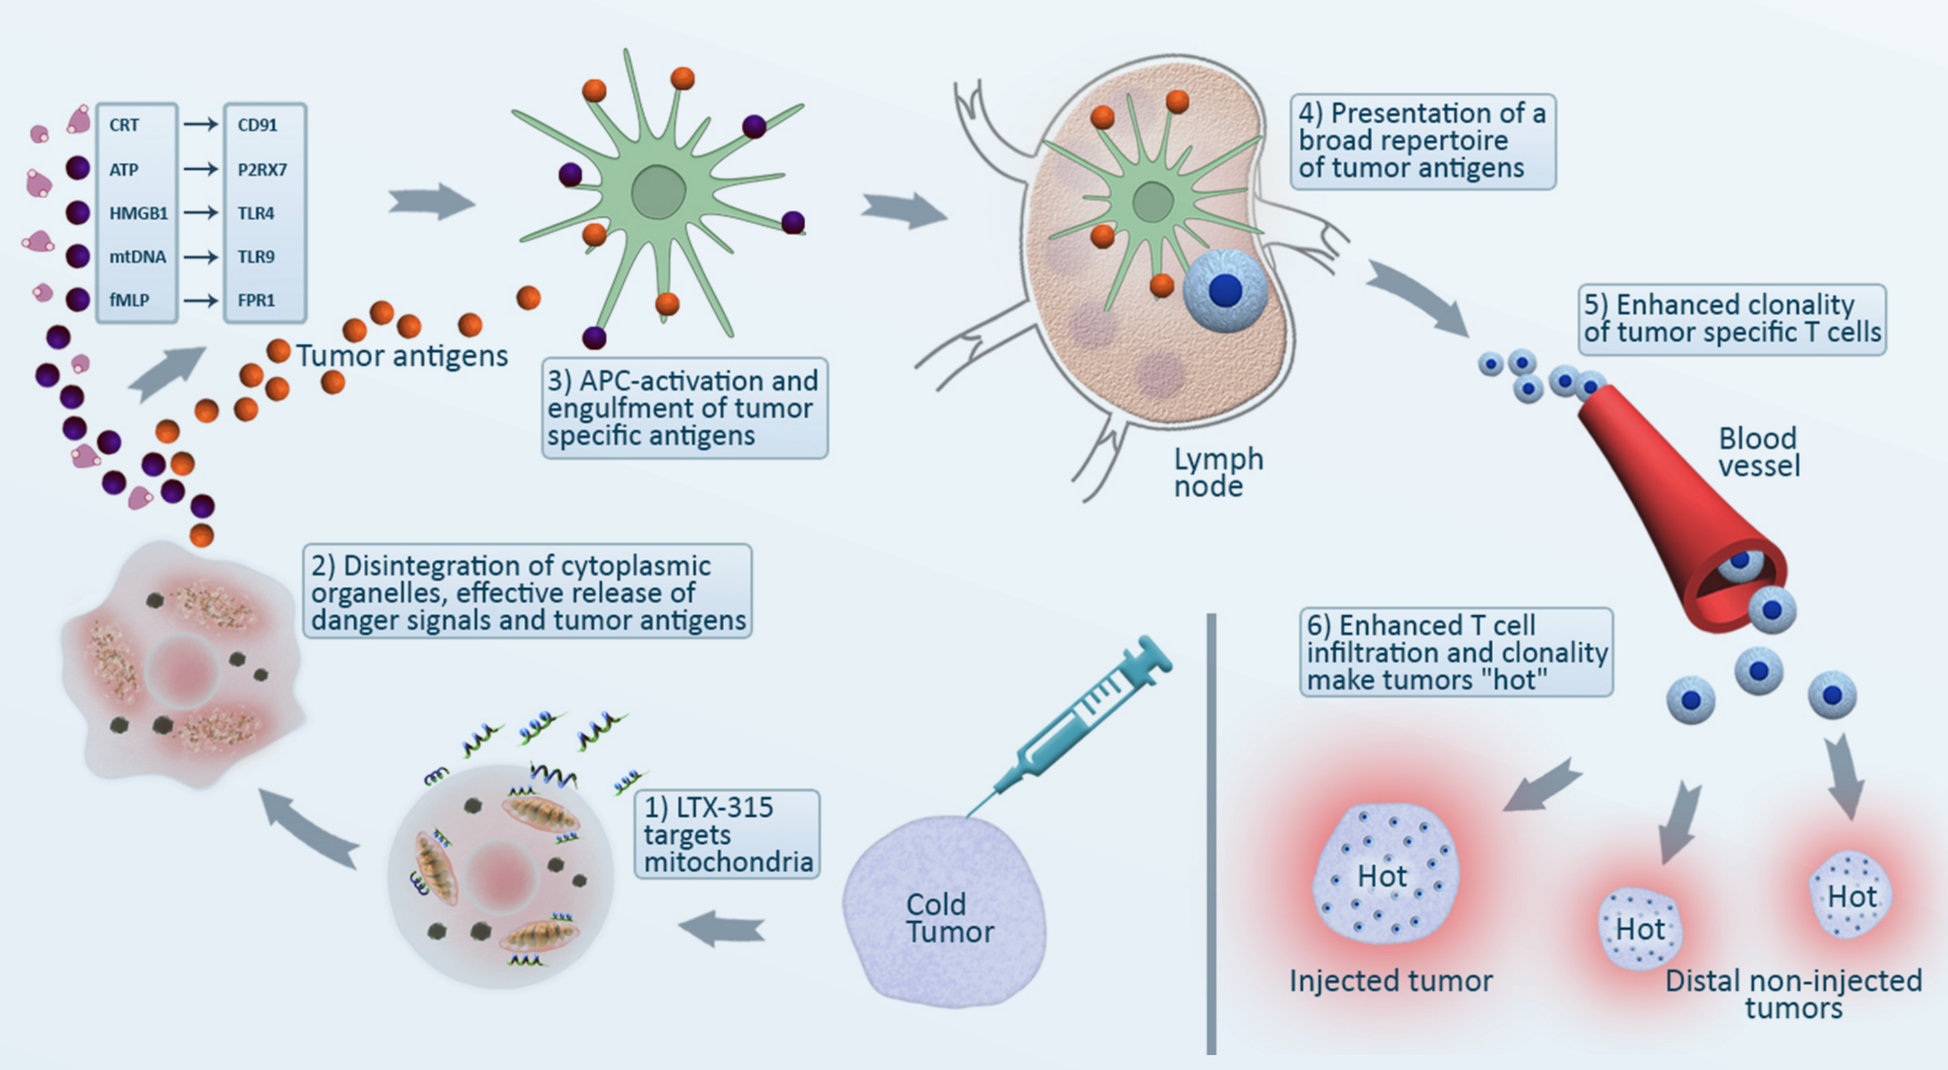
CRT
ATP
HMGB1
mtDNA
fMLP
CD91
P2RX7
TLR4
TLR9
FPR1
Tumor antigens
3) APC-activation and engulfment of tumor specific antigens
4) Presentation of a broad repertoire of tumor antigens
Lymph node
5) Enhanced clonality of tumor specific T cells
Blood vessel
2) Disintegration of cytoplasmic organelles, effective release of danger signals and tumor antigens
6) Enhanced T cell infiltration and clonality make tumors "hot"
1) LTX-315 targets mitochondria
Cold Tumor
Hot
Injected tumor
Distal non-injected tumors

LTX-315 demonstrates synergy with anti-CTLA4 or anti-PD-1 (4)

LTX-315 and anti-PD1 in murine sarcoma

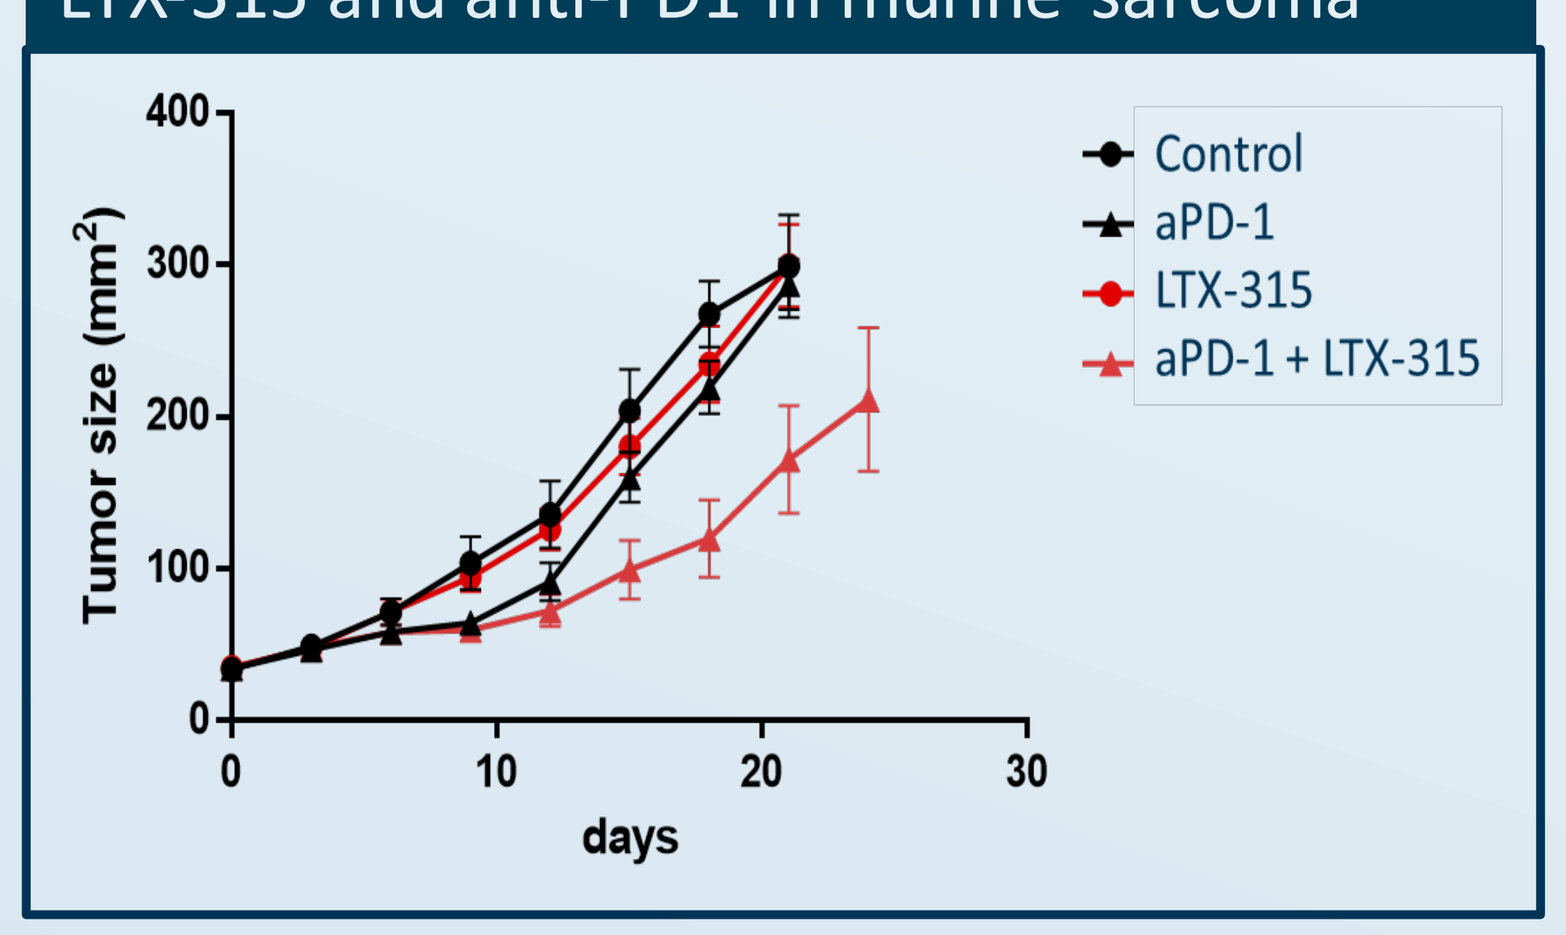
Tumor size (mm²)
days
Control
aPD-1
LTX-315
aPD-1 + LTX-315

LTX-315 and anti-CTLA-4 in murine sarcoma

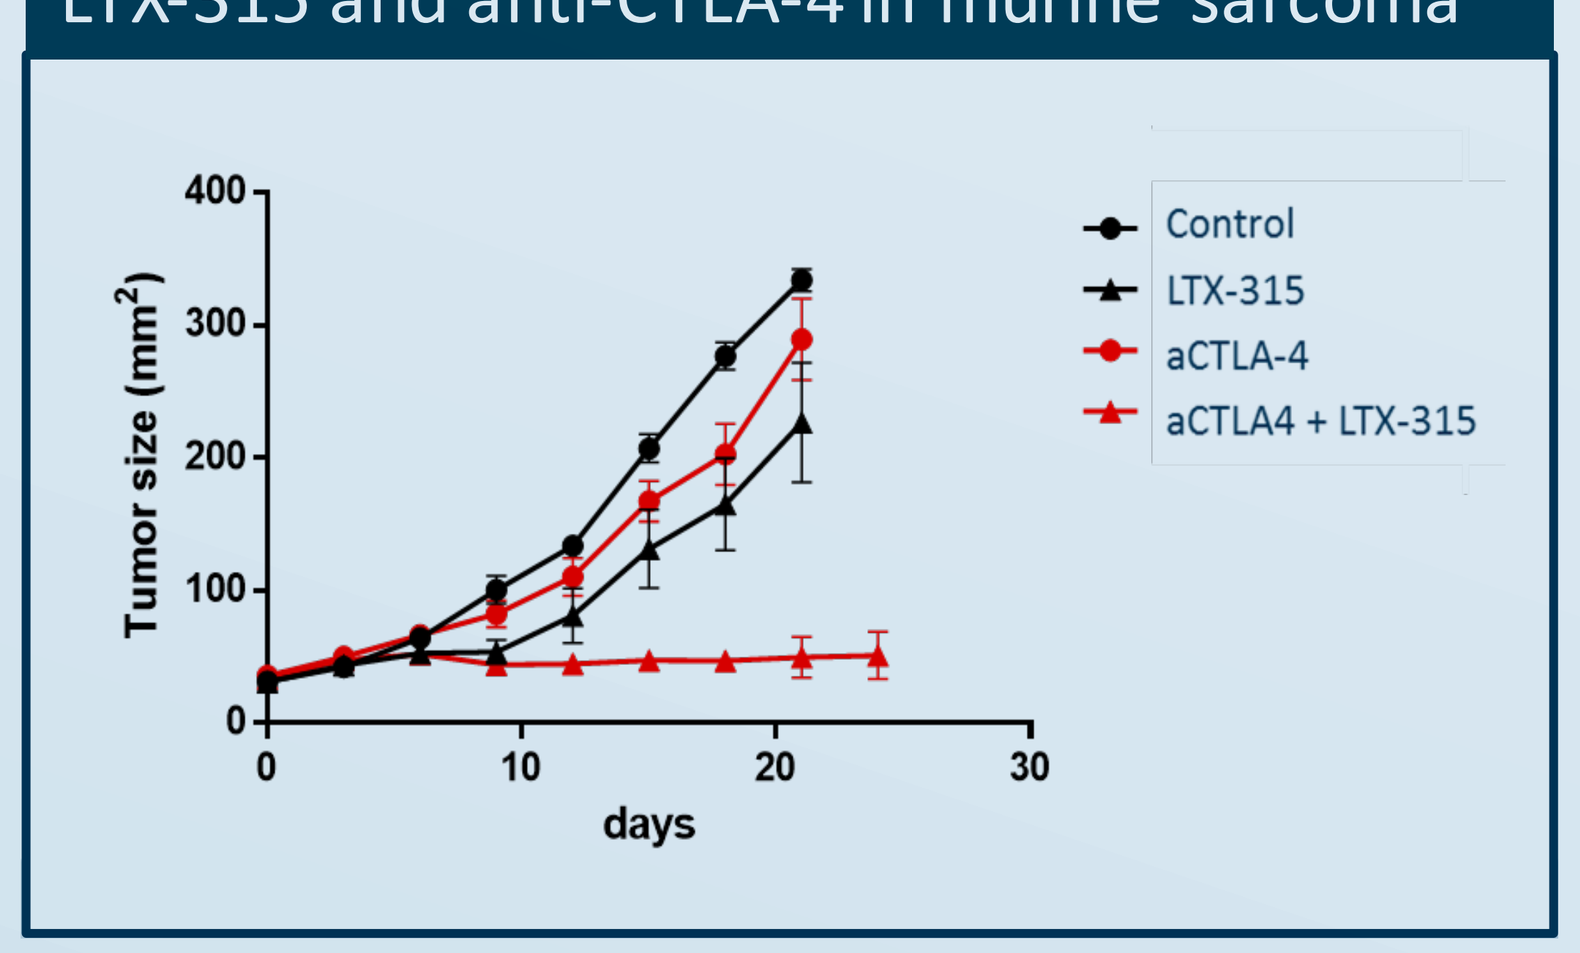
Tumor size (mm²)
days
Control
LTX-315
aCTLA-4
aCTLA4 + LTX-315

Aim

The aim of this study is to evaluate safety and tolerability of intratumoral doses of LTX-315 in patients with advanced/metastatic tumors in order to determine a recommended Phase II dose as a monotherapy and in combination with immune checkpoint inhibitors.

Study design

Arm A: Single lesion treatment arm

Patients with at least 1 injectable tumor lesion. LTX-315 injection follows a sequential schedule with each lesion treated for 6 weeks (induction treatment) followed by bi-weekly maintenance treatment

Arm A

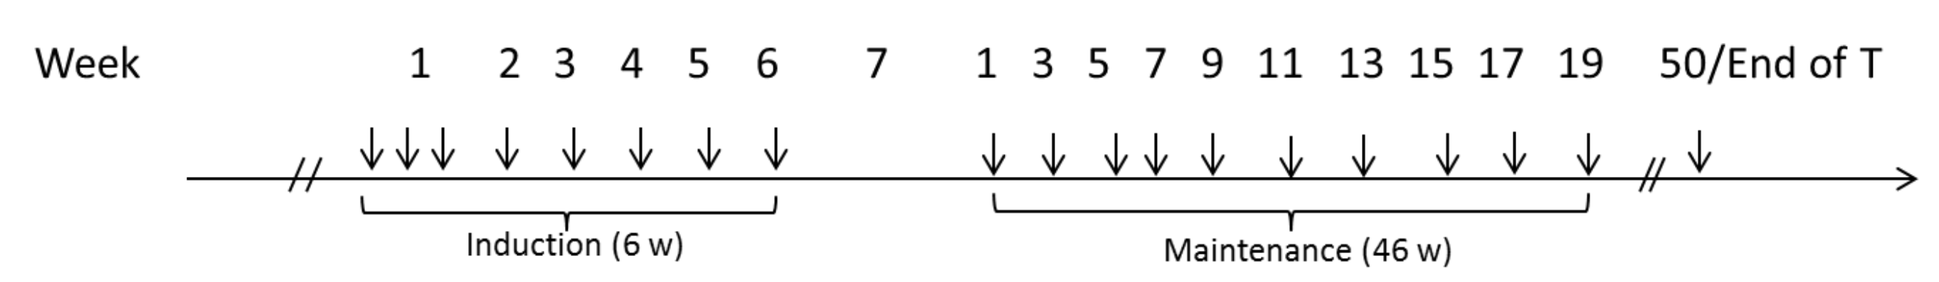
Week
1 2 3 4 5 6 7 1 3 5 7 9 11 13 15 17 19 50/End of T
Induction (6 w)
Maintenance (46 w)

Arm B: LTX-315 Monotherapy in patients with any solid tumor

Patients with different doses of LTX-315 starting with 3 mg. A minimum of one lesion is treated with a fixed dose of LTX-315 given as a single agent.

Arm C: LTX-315 in combination with ipilimumab in patients with unresectable/metastatic malignant melanoma

Patients with different doses of LTX-315, starting with 3 mg. In each cohort a minimum of one lesion is treated with a fixed dose of LTX-315 in combination with standard approved ipilimumab dose (3 mg/kg x 4 infusions).

Arm D: LTX-315 in combination with pembrolizumab in patients with unresectable/metastatic triple negative breast cancer

Patients with different doses of LTX-315, starting with 3 mg. In each cohort a minimum of one lesion is treated with a fixed dose of LTX-315 in combination with pembrolizumab dose 200 mg (every 21 days).

Arm B, C and D

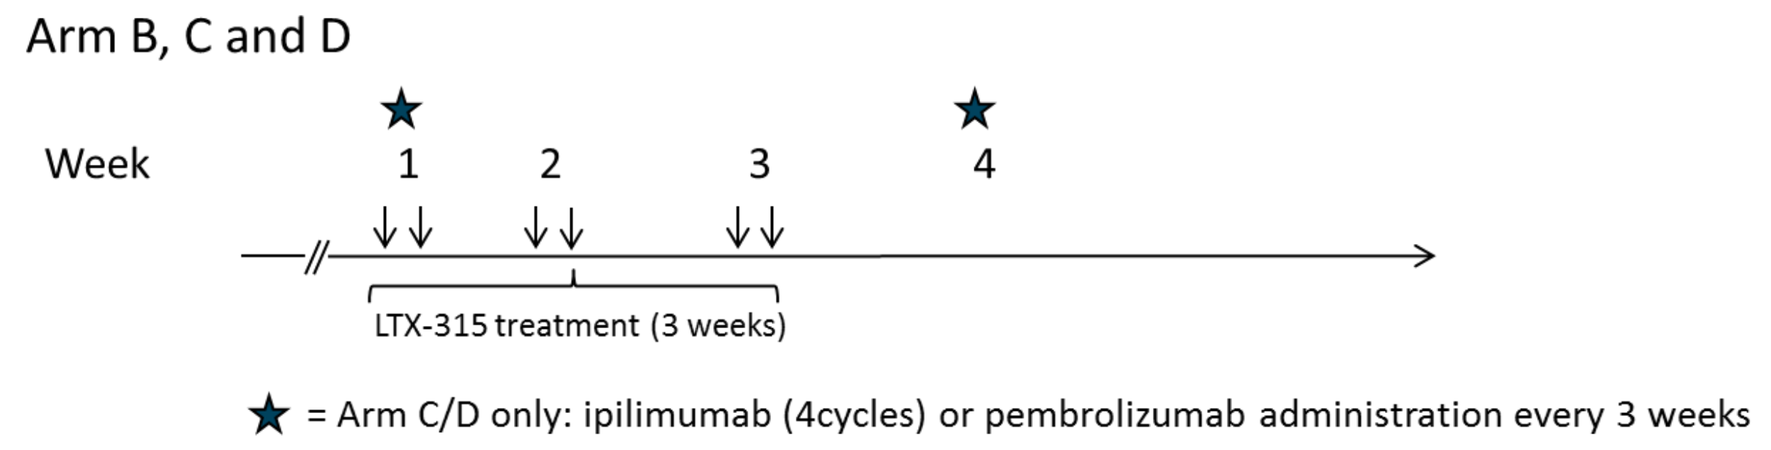
Week
1 2 3 4
LTX-315 treatment (3 weeks)
★ = Arm C/D only: ipilimumab (4cycles) or pembrolizumab administration every 3 weeks

Primary Endpoints

Dose limiting toxicities (DLT) and the overall safety profile of LTX-315 as a monotherapy, and in combination with ipilimumab or with pembrolizumab.

Secondary Endpoints

Immune monitoring:

- All arms: T lymphocyte subsets in systemic (peripheral blood) and injected tumors.
- Arm B: T Lymphocytes and PD-L1 expression in bystander (non-injected) tumor biopsies.

Anti-tumor activity of LTX-315 in injected tumor lesions as assessed by ultrasound and/or CT/MRI.

Anti-tumor activity assessed by the immune-related response criteria (irRC) for measureable lesions

- Response (complete response (irCR) and partial response (irPR)).
- Overall response duration, progression free survival (PFS), time to response.
- Disease control rate (irPR, irCR and stable disease (irSD)).

Treatment schedule (B, C, D)

Patients with at least one lesion available for injection receive LTX-315 (to all available lesions) as follows:

- Days 1, 2, 8, 9, 15 and 16 (Weeks 1-3)

The number of LTX-315 injections to a single lesion is depending on the volume of the lesion. Larger lesions may receive more than one injection. The total dose of LTX-315 will remain constant and the volume injected will be increased proportionally to allow for multiple injections.

The mean Longest perpendicular diameter (LPD) of a lesion will be used as a surrogate for assessing lesion volume.

LTX-315: Administered to all lesions available for injection at a minimum of 5 minutes apart and only if no grade 3, 4, or clinically significant or emergent (specifically allergy-like) adverse events have occurred or are emerging in the observation period between injections.

Starting LTX-315 dose for the multiple lesion cohort is 3 mg per injection. A maximum of 12 LTX-315 injections per day is allowed in 1 hour depending on lesion volume.

Pembrolizumab: Every 21 days starting on Day 2 in Week 1 until progressive disease (PD) or unacceptable toxicity

Ipilimumab: Every 21 days starting on Day 2 in Week 1 for 4 cycles

Inclusion criteria

Arm A/B:

- Unresectable advanced or metastatic disease (**any tumor type**) and previously received treatment with all available standard of care treatments or are not eligible or suitable for such treatments.
- At least one available lesion (cutaneous, sub-cutaneous or lymph node) for injection which is between 1 and 3 cm in longest diameter, and one bystander (non-injected) lesion.

Arm C:

- Unresectable/metastatic diagnosis of **malignant melanoma** (histologically confirmed).
- At least one available lesion (cutaneous, sub-cutaneous, oral or lymph node) for injection and biopsy which is between 1 and 3 cm in longest diameter.
- Previous treatment with an anti-PD-1 antibody (as monotherapy or as part of combination (any combination) as 1st or 2nd line metastatic treatment).

Arm D:

- Unresectable/metastatic diagnosis of **triple negative breast** cancer (histologically confirmed).
- At least one available lesion (cutaneous, sub-cutaneous or lymph node) for injection and biopsy with a minimum longest diameter of 1 cm.
- Received between one and 4 prior systemic treatments for metastatic triple negative breast cancer.

Exclusion criteria

Arm B:

- Received cancer immunotherapy within 2 weeks prior to study drug administration or have not recovered from adverse events (to ≤ CTCAE grade 1) due to such agents.
- History of systemic auto-immune disease requiring anti-inflammatory or immunosuppressive therapy within the last 3 months. Patients with history of autoimmune thyroiditis are eligible provided the patient requires only thyroid hormone replacement therapy and disease has been stable for ≥ 1 year.

Arm C:

- Prior therapy with ipilimumab or any other anti-CTLA-4 monoclonal antibody.
- Known hypersensitivity to any of the excipients in ipilimumab infusion
- Received cancer immunotherapy within 2 weeks prior to study drug administration or have not recovered from adverse events (to ≤ CTCAE grade 1) due to such agents.
- Had BRAF/MEK inhibitors administered within 2 weeks prior to the study drug administration.

Arm D:

- Prior therapy with an anti-PD-1 or anti-PD-L1 monoclonal antibody.
- Known hypersensitivity to any of the excipients pembrolizumab infusion.
- Received cancer immunotherapy within 2 weeks prior to study drug administration or have not recovered from adverse events (to ≤ CTCAE grade 1) due to such agents.

All arms:

- Active systemic autoimmune disease; prior pneumonitis; history of severe hypersensitivity to another monoclonal antibody; receiving immunosuppressive therapy; history of severe immune-related adverse reaction from treatment with a monoclonal antibody, defined as any Grade 4 or 3 toxicity requiring corticosteroid treatment (> 10 mg/day prednisone or equivalent) for greater than 12 weeks.

Biopsy/Resection schedules

- All mandatory biopsies timepoints include up to three core biopsies.
- **Arm A:** One or more injected lesions are biopsied prior to LTX-315 day 1 administration, Day 8 and at Follow up
- **Arm B: injected lesions** - One or more lesions available for injection are biopsied prior to LTX-315 administration Day 1 and Week 4. **Bystander lesions** - one or more bystander lesions are biopsied prior to LTX-315 Day 1 administration, Week 8 and at Follow-up. At least one bystander lesion is surgically excised (mandatory) at the end of study or at discontinuation. This will facilitate confirmation of a systemic immune response.
- **Arm C and D: injected lesions** - One or more lesions available for injection are biopsied prior to LTX-315 administration and at Week 4

Recommended Phase II dose

The decision on the optimal phase II LTX-315 treatment regimen will be based on the results of the dose escalation cohorts from the following information:

- Safety parameters (DLTs, AEs etc.).
- Systemic inflammatory response such as total lymphocyte count.
- Clinical efficacy: Overall Response Rate (ORR), Stable Disease (SD), Clinical Benefit Rate (CBR) (irRC criteria).

Patient characteristics	
Median age (range)	59 (32-80)
Male: Female	11:17
Tumor type	No. of patients
Breast	7
Melanoma	7
Head and neck	4
Other	10
ECOG PS	No. of patients
0	8
1	20
No. of prior treatments for advanced/metastatic disease	No. of patients
Median (range)	3(0-20)
0	2
1-2	10
3-4	8
≥ 5	8
Median number of LTX-315 injections (range)	14 (4-54)
LTX-315 dose	**No of injected lesions**
2mg BD	1 – 2
3mg BD	2 – 3
4mg BD	1 – 2
4→5mg QD	1
4→6mg QD	1
4→7mg QD	1
3mg in each lesion	3 – 6
4mg in each lesion	1 – 3

Conclusion

- LTX-315`s unique "release and reshape" properties makes it ideal for combination with other types of immunotherapy.
- The preclinical results with LTX-315 in combination with anti-CTLA-4 and anti-PD-1 confirms the scientific rational for combining LTX-315 with immune-checkpoint inhibitors in a clinical study.
- As per January 2017, 28 of planned 60 patients have been recruited to the study

References

1. Hancock & Sahl, Nature Biotechnology (2006)
2. Camilio et al. Cancer Immunol Immunother (2014)
3. Source: data on file, manuscript in preparation
4. Yamazaki, Cell Death and Differentiation. 2016

CHILI. @WWW.CHILIGROUP.NO | 2017

Lytix Biopharma AS | Gaustadalléen 21 | 0349 Oslo, Norway | E-mail: post@lytixbiopharma.com