560

Immunotherapeutic and antimetastatic activity of LTX-315 in preclinical models of ICI-resistant breast cancer

Takahiro Yamazaki[1] , Erik Wennerberg[1], Michal Hensler[2], Aitziber Buqué[1], Jeffrey Kraynak[1], Jitka Fucikova[2,3], Xi Kathy Zhou[4], Baldur Sveinbjørnsson[5,6,7], Øystein Rekdal[5,6], Sandra Demaria[1,8], Lorenzo Galluzzi[1,8,9]

[1]Department of Radiation Oncology, Weill Cornell Medical College, New York, NY, USA; [2]Sotio, Prague, Czech Republic; [3]2nd Faculty of Medicine and University Hospital Motol, Department of Immunology, Charles University, Prague, Czech Republic; [4]Department of Population Health Sciences, Weill Cornell Medical College, New York, NY, USA; [5]Lytix Biopharma, Oslo, Norway; [6]Department of Medical Biology, University of Tromsø, Tromsø, Norway; [7]Childhood Cancer Research Unit, Department of Women and Children Health, Karolinska Institute, Stockholm, Sweden; [8]Sandra and Edward Meyer Cancer Center, New York, NY, USA; [9]Caryl and Israel Englander Institute for Precision Medicine, New York, NY, USA

Introduction

Oncolytic peptides are attractive tools for the development of novel anticancer regimens [1]. LTX-315 is a synthetic peptide with a marked capacity to elicit tumor-targeting immunity in preclinical cancer models [2]. Indeed, LTX-315 has been shown to elicit immunogenic cell death (ICD) in malignant cells [3, 4] and to deplete immunosuppressive cells such as $CD4^+CD25^+FOXP3^+$ T_{REG} cells and myeloid-derived suppressor cells (MDSCs) from the tumor microenvironment (TME) [5]. Accordingly, LTX-315 synergized with immunogenic chemotherapeutics or immune checkpoint inhibitors (ICIs) in preclinical tumor models [5, 6]. Moreover, recent findings from a Phase I clinical trial in patients with advanced solid tumors (NCT01986426) indicate that intratumoral LTX-315 is safe, clinically active, and elicits alterations in the TME that support the initiation of anticancer immunity [7, 8]. However, the dependency of LTX-315 therapeutic effects on the immune system in preclinical models of breast cancer has not been mechanistically investigated. Here, we examined the ability of LTX-315 to cooperate with a panel of common immunostimulatory agents in mouse models of hormone receptor (HR)+ and triple-negative breast cancer, focusing on the immunological correlates of activity and underlying mechanisms of action. Our findings suggest that LTX-315 controls breast cancer progression by immunotherapeutic effects that (at least in some settings) can be boosted by radiation therapy (RT), persist in the context of ICI-based immunotherapy, and rely on natural killer (NK) cells.
This work **"LTX-315-enabled, radiotherapy-boosted immunotherapeutic control of breast cancer by NK cells"** has been published in Oncoimmunology (2021).

References

1. Kepp, O. et al. (2020) Oncolysis without viruses - inducing systemic anticancer immune responses with local therapies. Nat Rev Clin Oncol 17 (1), 49-64.

2. Vitale, I. et al. (2021) Targeting Cancer Heterogeneity with Immune Responses Driven by Oncolytic Peptides. Trends Cancer 7 (6), 557-572.

3. Eike, L.M. et al. (2015) The oncolytic peptide LTX-315 induces cell death and DAMP release by mitochondria distortion in human melanoma cells. Oncotarget 6 (33), 34910-23.

4. Zhou, H. et al. (2016) The oncolytic peptide LTX-315 triggers immunogenic cell death. Cell Death Dis 7 (3), e2134.

5. Yamazaki, T. et al. (2016) The oncolytic peptide LTX-315 overcomes resistance of cancers to immunotherapy with CTLA4 checkpoint blockade. Cell Death Differ 23 (6), 1004-15.

6. Camilio, K.A. et al. (2019) Combining the oncolytic peptide LTX-315 with doxorubicin demonstrates therapeutic potential in a triple-negative breast cancer model. Breast Cancer Res 21 (1), 9.

7. Jebsen, N.L. et al. (2019) Enhanced T-lymphocyte infiltration in a desmoid tumor of the thoracic wall in a young woman treated with intratumoral injections of the oncolytic peptide LTX-315: a case report. J Med Case Rep 13 (1), 177.

8. Spicer, J. et al. (2021) Safety, Antitumor Activity, and T-cell Responses in a Dose-Ranging Phase I Trial of the Oncolytic Peptide LTX-315 in Patients with Solid Tumors. Clin Cancer Res 27 (10), 2755-2763.

Immunotherapeutic control of triple-negative 4T1 mammary carcinomas by LTX-315

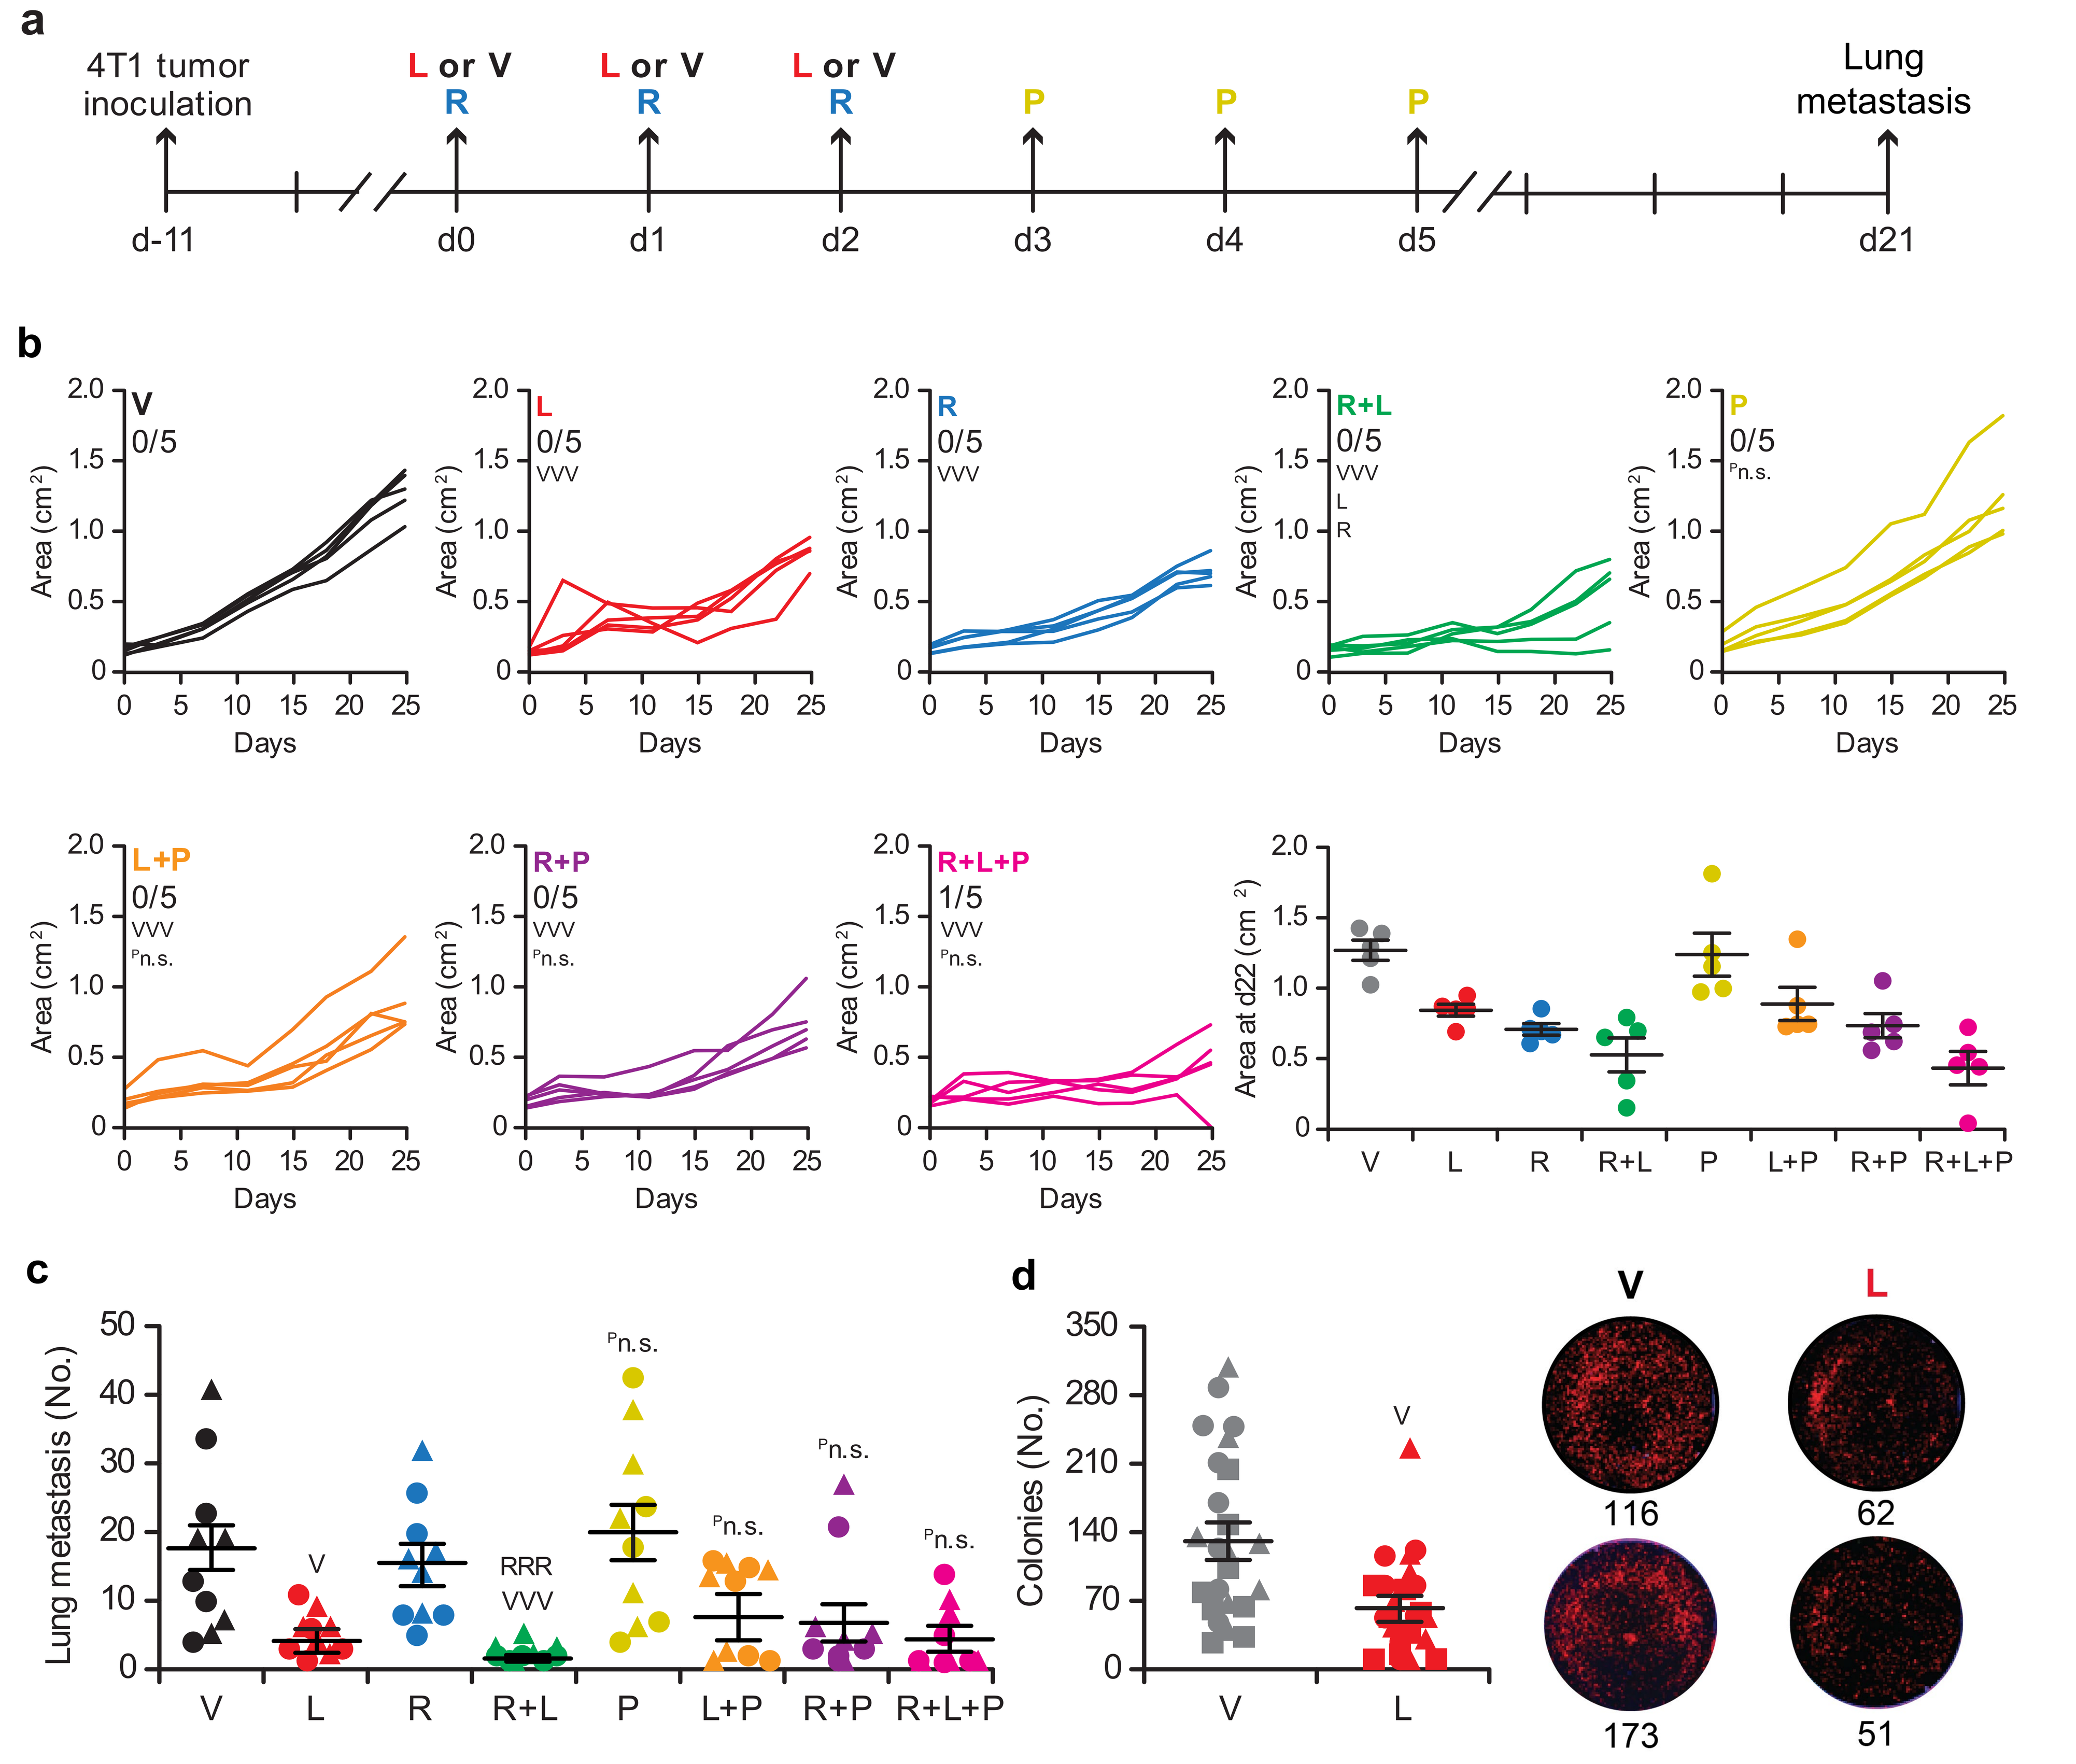
a
4T1 tumor inoculation
L or V R
P
Lung metastasis
d-11 d0 d1 d2 d3 d4 d5 d21
b
V 0/5
L 0/5 VVV
R 0/5 VVV
R+L 0/5 VVV L R
P 0/5 Pn.s.
L+P 0/5 VVV Pn.s.
R+P 0/5 VVV Pn.s.
R+L+P 1/5 VVV Pn.s.
Area (cm²)
Days
Area at d22 (cm²)
V L R R+L P L+P R+P R+L+P
c
Lung metastasis (No.)
V RRR VVV Pn.s.
d
Colonies (No.)
V L
116 62 173 51

(a) Experimental setup. L, LTX-315; (300 µg, *i.t.*) R, radiation (8 Gy); P, PD-1 blocker (RMP1-14, 200 µg, *i.p.*); V, vehicle.

(e) Growth of primary 4T1 mammary carcinomas established in BALB/c mice that were subjected to the local or systemic treatments illustrated in (a). Individual growth curves, incidence of tumor eradication as well as mean tumor area at d21 ± SEM and individual data points are reported. Vp < .05, VVVp < .001 (linear mixed-effects model plus simultaneous tests for general linear hypotheses, Wilcoxon rank sum test for tumor area), as compared to V-treated mice; Rp < .05, as compared to R-treated mice; Lp < .05, as compared to L-treated mice; Pn.s., not significant, as compared to mice receiving the same treatment in the absence of PD-1 blockage.

(f) Number of macroscopic lung metastases in BALB/c mice bearing 4T1 mammary carcinomas that were subjected to the local or systemic treatments illustrated in (a). Results are means ± SEM and individual data points from two independent operators. Vp < .05, VVVp < .001 (linear mixed-effects model plus simultaneous tests for general linear hypotheses), as compared to V-treated mice; RRRp < .001, as compared to R-treated mice; Pn.s., not significant, as compared to mice receiving the same treatment in the absence of PD-1 blockage.

(g) Number of colony-forming cells isolated from the lung of 4T1-bearing BALB/c mice treated with vehicle or LTX-315 as illustrated in (a). Results are means ± SEM and individual data points from two independent operators plus representative images from clonogenic assays. Vp < .05 (Welch test), as compared to V-treated mice.

NK cells as to central players in the ability of LTX-315 to control breast cancer progression

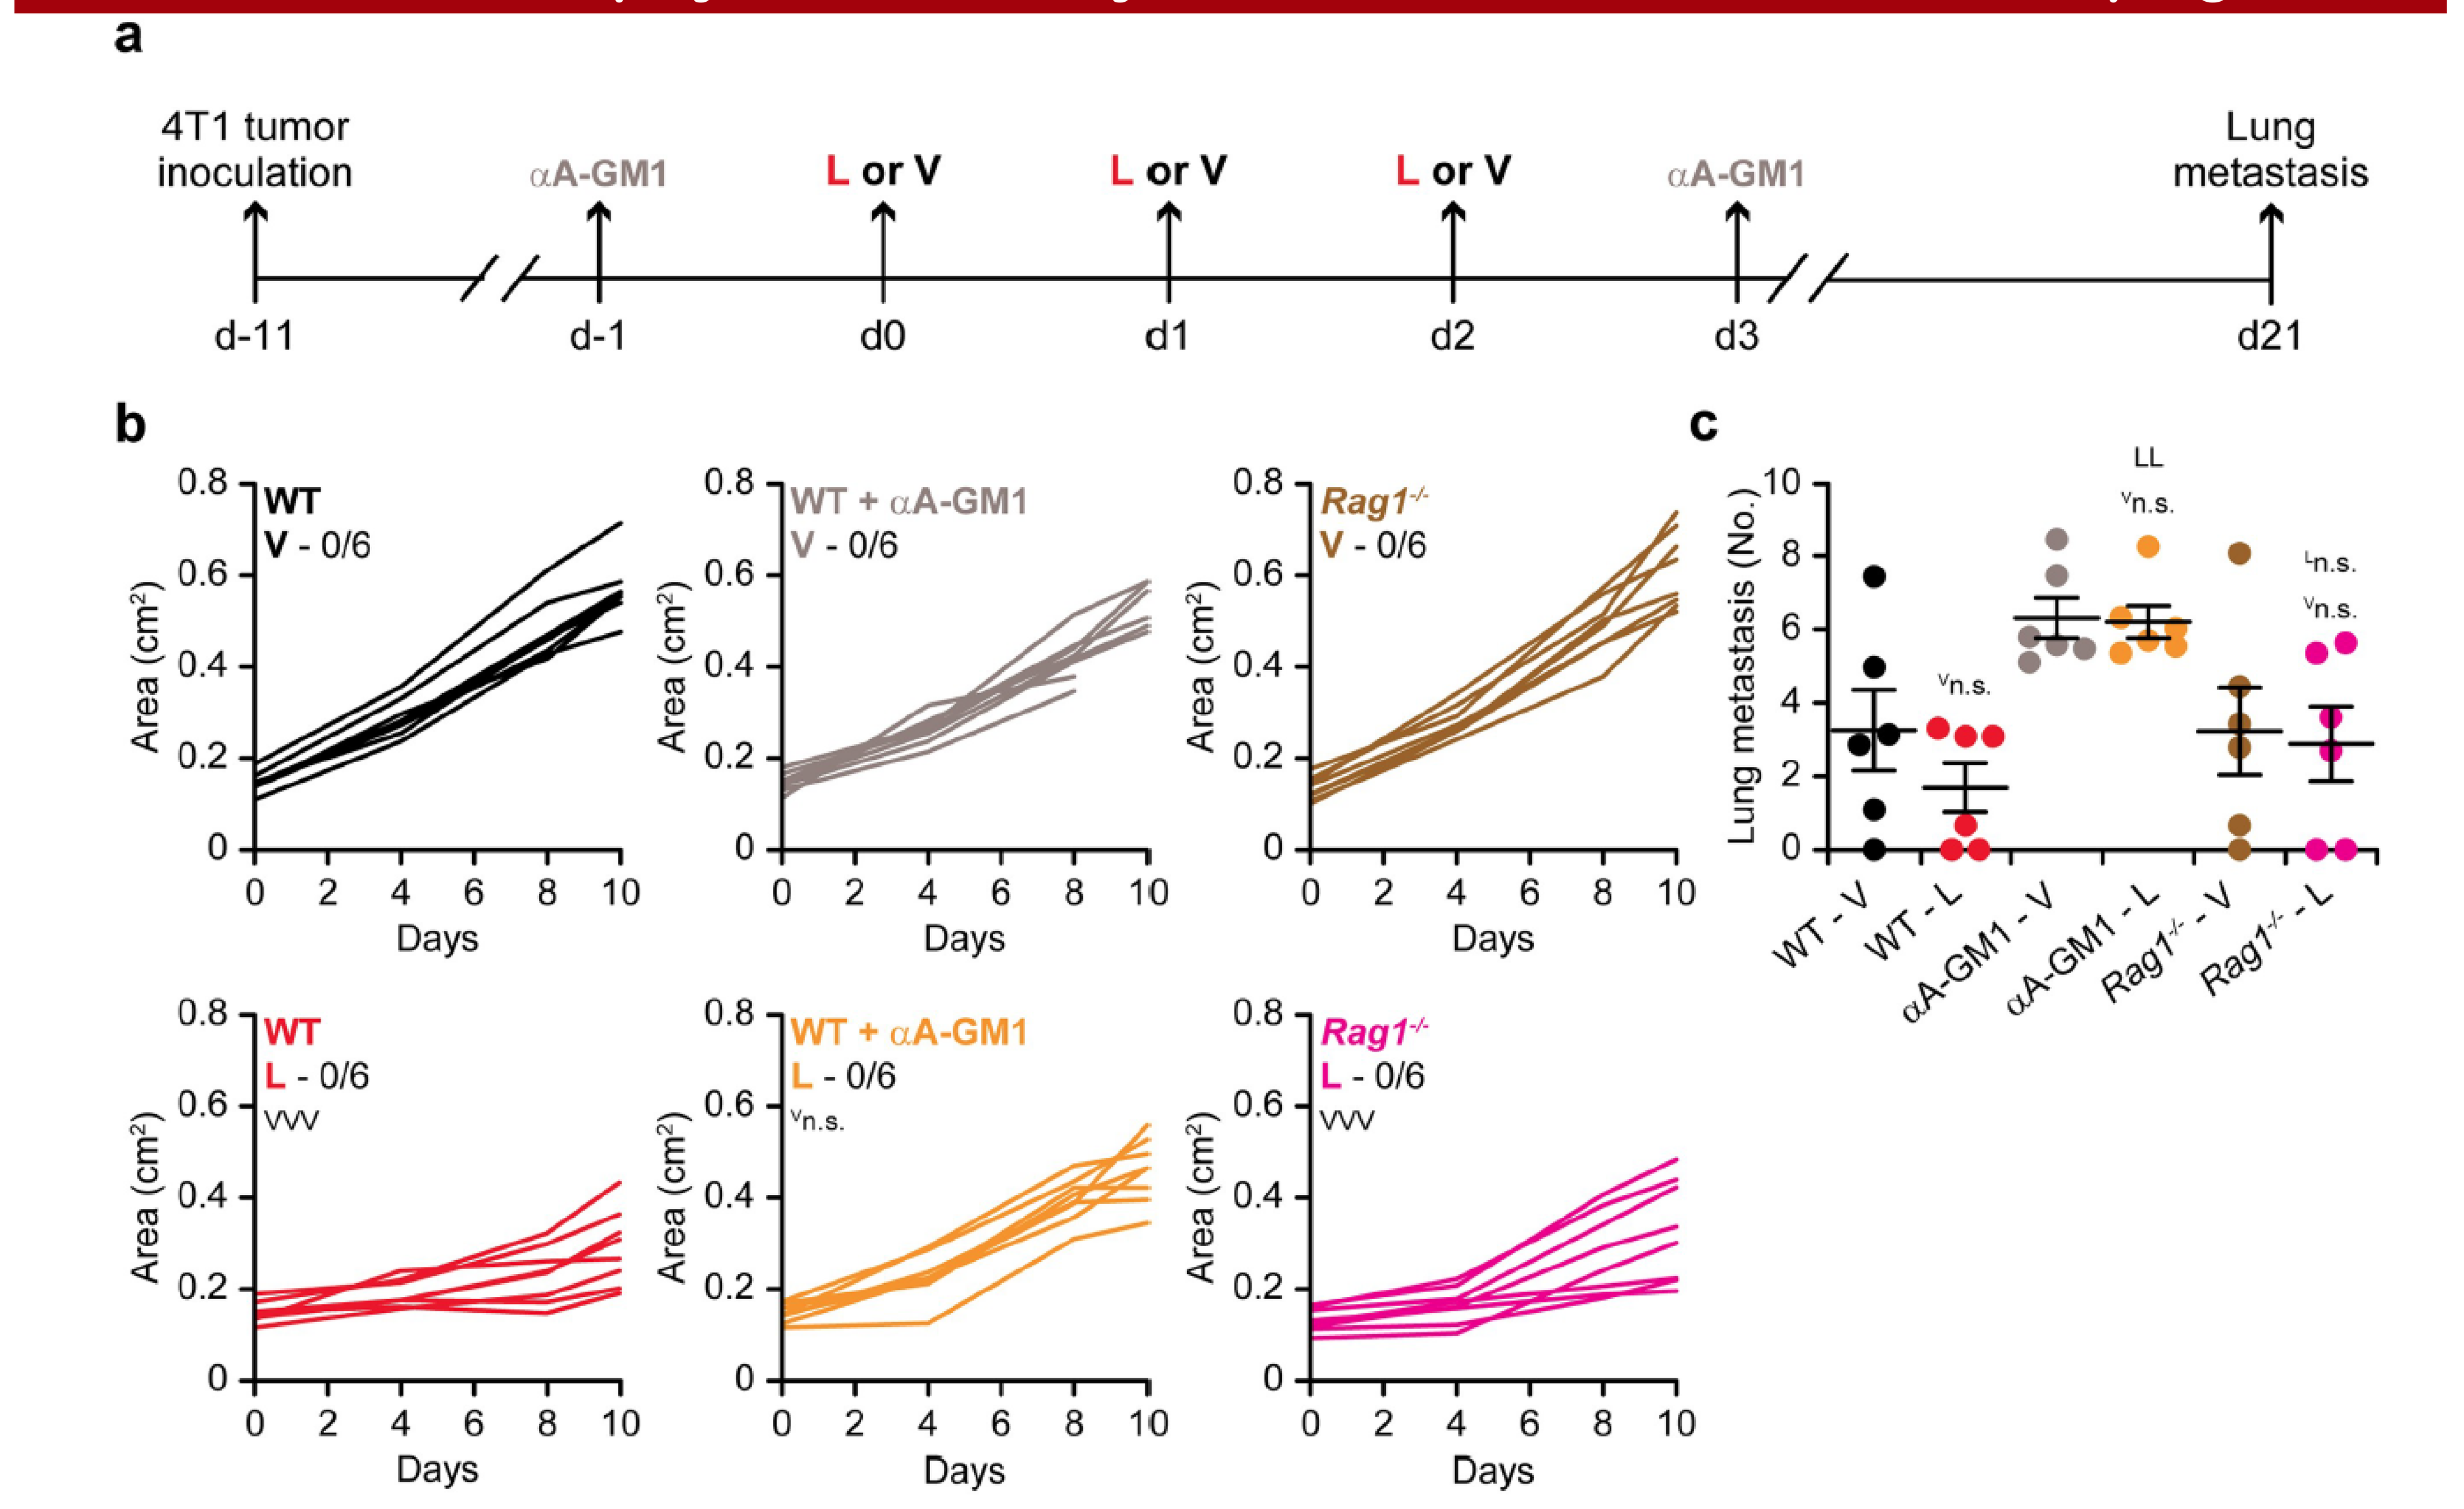
a
4T1 tumor inoculation
αA-GM1
L or V
Lung metastasis
d-11 d-1 d0 d1 d2 d3 d21
b
WT V - 0/6
WT + αA-GM1 V - 0/6
Rag1-/- V - 0/6
WT L - 0/6 VVV
WT + αA-GM1 L - 0/6 Vn.s.
Rag1-/- L - 0/6 VVV
Area (cm²)
Days
c
Lung metastasis (No.)
LL Vn.s. Ln.s.
WT - V WT - L αA-GM1 - V αA-GM1 - L Rag1-/- - V Rag1-/- - L

(a) Experimental setup for 4T1 tumors. αA-GM1, asialo GM1-targeting antibody; LTX-315; (300 µg, *i.t.*); V, vehicle.

(b) Growth of primary 4T1 mammary carcinomas established in wild-type (WT) or *Rag1−/−* BALB/c mice that were subjected to the local or systemic treatments illustrated in (a). Individual growth curves and incidence of tumor eradication are reported. Vn.s., not significant, VVVp < .001 (linear mixed-effects model plus simultaneous tests for general linear hypotheses) as compared to V-treated mice of the same genotype and subjected to the same antibody-mediated depletion regimen.

(c) Number of macroscopic lung metastases in WT or *Rag1−/−* BALB/c mice bearing 4T1 mammary carcinomas that were subjected to the local or systemic treatments illustrated in (a). Results are means ± SEM and individual data points from a single operator. Vn.s., not significant (Wilcoxon rank sum test), as compared to V-treated mice of the same genotype and subjected to the same antibody-mediated depletion regimen; Ln.s., not significant, LLp < .01 (Wilcoxon rank sum test), as compared to L-treated WT mice.

Conclusions

In 4T1 triple-negative breast cancer model, PD-1 had no effect on primary tumor growth but LTX-315 remained active in the context of ICI-based immunotherapy, both when employed alone and when combined with RT. Consistent with local disease control, LTX-315 caused a considerable decrease in the number of macroscopic pulmonary metastases formed by progressing 4T1 tumors, an effect that was marginally improved by RT and persisted in the presence of PD-1 blockers.

From results of Rag KO and NK depletion experiments, mechanistically point to NK cells as to central players in the ability of LTX-315 to control breast cancer progression and metastasis (locally and systemically), potentially in the context of memory-like responses. Thus, the ability of LTX-315 to control metastatic dissemination may be further boosted by interventions aimed at enhancing systemic NK cell functions such as (1) adoptive NK cell transfer, (2) recombinant IL15 administration, or (3) killer cell lectin like receptor C1 (KLRC1, best known as NKG2A) blockage.